Poster #5086
6/12/22

Automated colony picking and McFarland preparation using the Copan Colibri instrument is equivalent to manual McFarland preparations for antimicrobial susceptibility testing using the Vitek 2 system

K. Bryant, Kaiser Permanente, Portland OR

Kendall.A.Bryant@kp.org

ABSTRACT:

Introduction: The Copan Colibri instrument is an *in vitro* diagnostic device that automatically picks colonies from predetermined plate locations and prepares 0.5 McFarland standard suspensions for use with antimicrobial susceptibility testing (AST). The system also applies barcode labels to tubes and prepares purity check plates from the McFarland suspensions. This study used the Vitek2 (V2) AST system to compare McFarland suspensions prepared by the Colibri to those prepared manually. **Methods:** 106 organisms were compared in the study (56 GNR, 50 GPC) using V2 GN81 and GP75 panels. The organisms were comprised of clinical isolates and reference organisms from the CDC AR Isolate Bank. V2 results were used as the comparator result for the clinical isolates and the known MICs of the CDC organisms were used as the comparator result for the banked isolates. **Results:** 1732 drug/bug combinations were tested with the 106 organisms. There were 6 discrepant results in categorical interpretations with the clinical isolates: 1) gentamicin-R (manual) and -I (Colibri), and 2) nitrofurantoin-I (manual) and -R (Colibri), and 1) amoxicillin/clavulanate I (manual) and -R (Colibri) and 1) nitrofurantoin-S (manual) and -R (Colibri) and1) and there was 1 discrepant result with a CDC organism where the Colibri suspension agreed with the CDC result of S while the manual result was I. This resulted in an overall categorical agreement of <u>99.99%</u>. There were 5 instances of > 1 dilution difference in MIC results between the two methods: 1) cefazolin with a K*lebsiella pneumoniae* clinical isolate, 1) ampicillin and E*scherichia coli* clinical isolate, 1) erythromycin with a *Staphylococcus hominis* and 2) daptomycin with a *Staphylococcus aureus* clinical isolate for an overall +/- 1 dilution correlation of <u>99.99%</u>. **Conclusions:** The Colibri System's performance is equivalent to manual methods for McFarland suspension preparation used for susceptibility testing on the V2.

INTRODUCTION:

The Copan Colibrí (**Figure 1**) is an instrument that works in concert with WASPLab to further automate manual microbiological processes used for identification and susceptibility testing of clinical isolates. The Colibri automates procedures for susceptibility testing by making the 0.5 McFarland standard and subsequent dilution tubes needed by automated AST systems. The Colibrí also can prepare purity plates from the AST tubes.

The following study compares the use of the Colibrí to the standard manual method of preparing Vitek 2 AST cards. The Colibrí uses an onboard nephelometer to make a 0.5 McFarland suspension. Once the 0.5 McFarland is made, the Colibrí prepares the appropriate dilution in a secondary tube, labels this tube for Vitek 2 susceptibility testing, and prepares the purity plate.

MATERIALS AND METHODS

Isolates for comparison were collected from routine culture types such as urines, wounds, and respiratory specimens, and blood cultures which were plated and incubated in the WASPLab. The plates were incubated for 18-24 hours before being assigned 'pick points' (**Figure 2**) which indicate to the Colibrí which colonies to sample for AST. Once pick points were assigned to the culture plates, they were transferred to the Colibrí for Vitek AST preparation (**Figure 3**). The Colibrí sampled each of the pre-selected colonies with a pipette tip (**Figure 4**), transferred the colony to a preloaded primary tube, and checked the turbidity to ensure a 0.5 McFarland standard was prepared and made a dilution tube for Vitek 2 AST (**Figure 5**). A purity plate was inoculated and streaked from each of the secondary tubes (**Figure 6**). The secondary tubes were used to inoculate either a Vitek AST-GN81 card for Gram negative organisms or a Vitek AST-GP75 card Gram positive organisms. The results of the Colibrí AST suspensions were compared to the manual made 0.5 McFarland standard preparations using the Vitek DensiChek device and the standard Vitek 2 setup protocol. A total of 156 isolates were compared in the study 56 Gram negative organisms and 50 Gram positive organisms.

Figure 1: Copan's Colibrí instrument

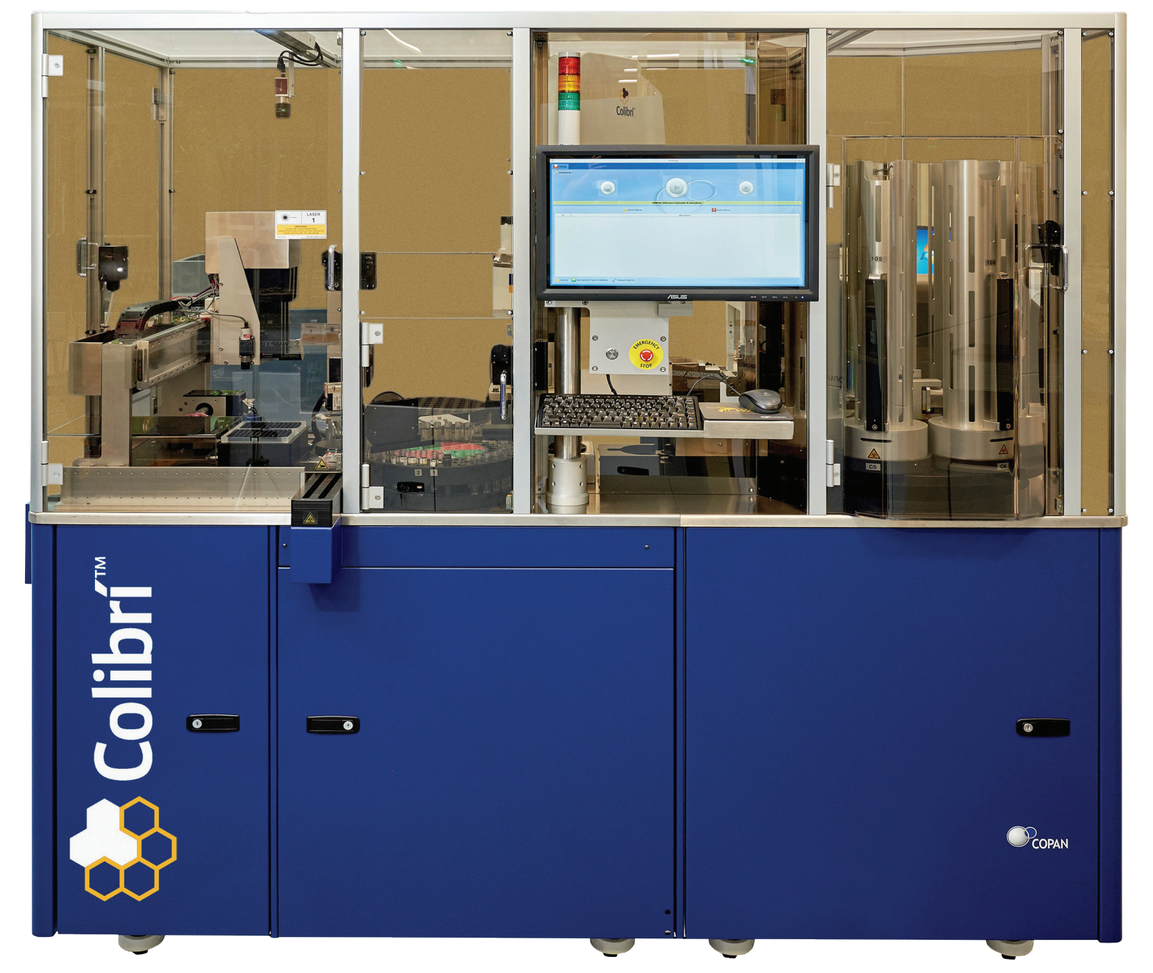
Colibri

Figure 2: Pick points selected by technologists in the WASPLab Webapp.

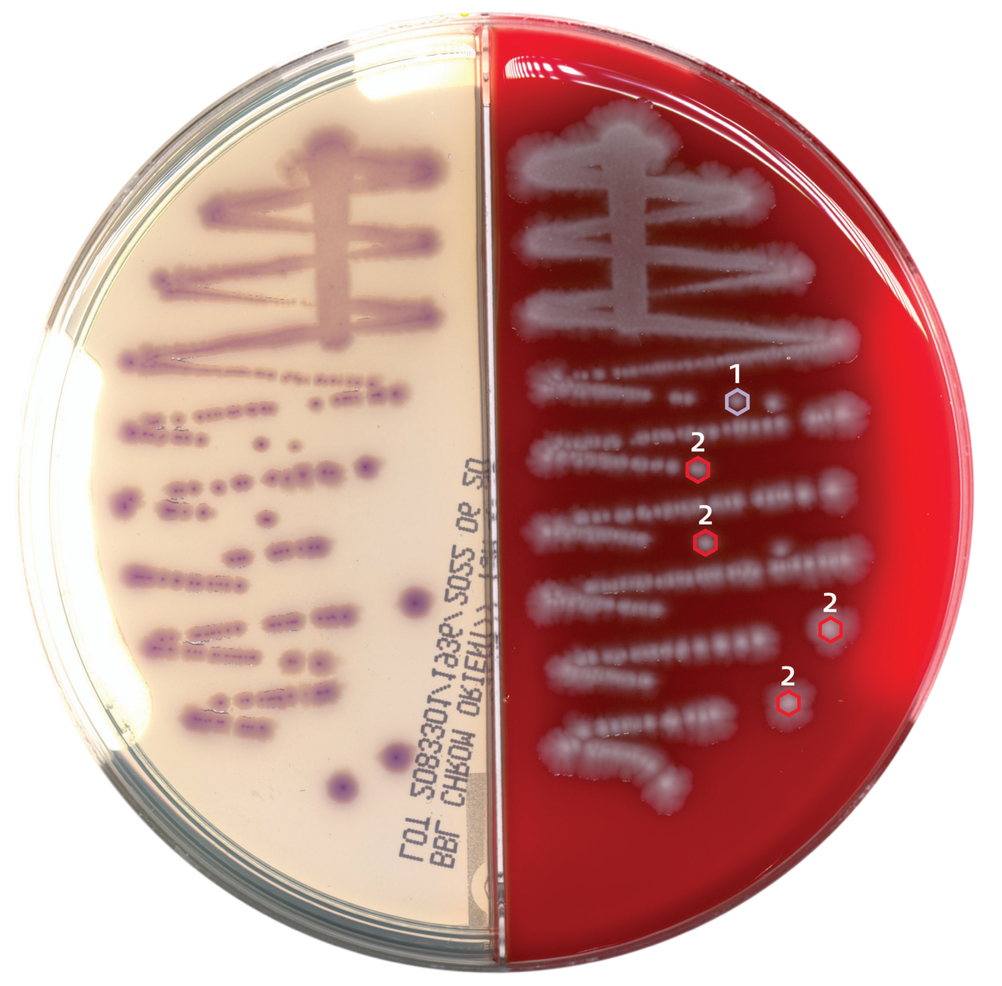

Figure 3: Canisters with pick point-culture plates are delivered to the Colibrí.

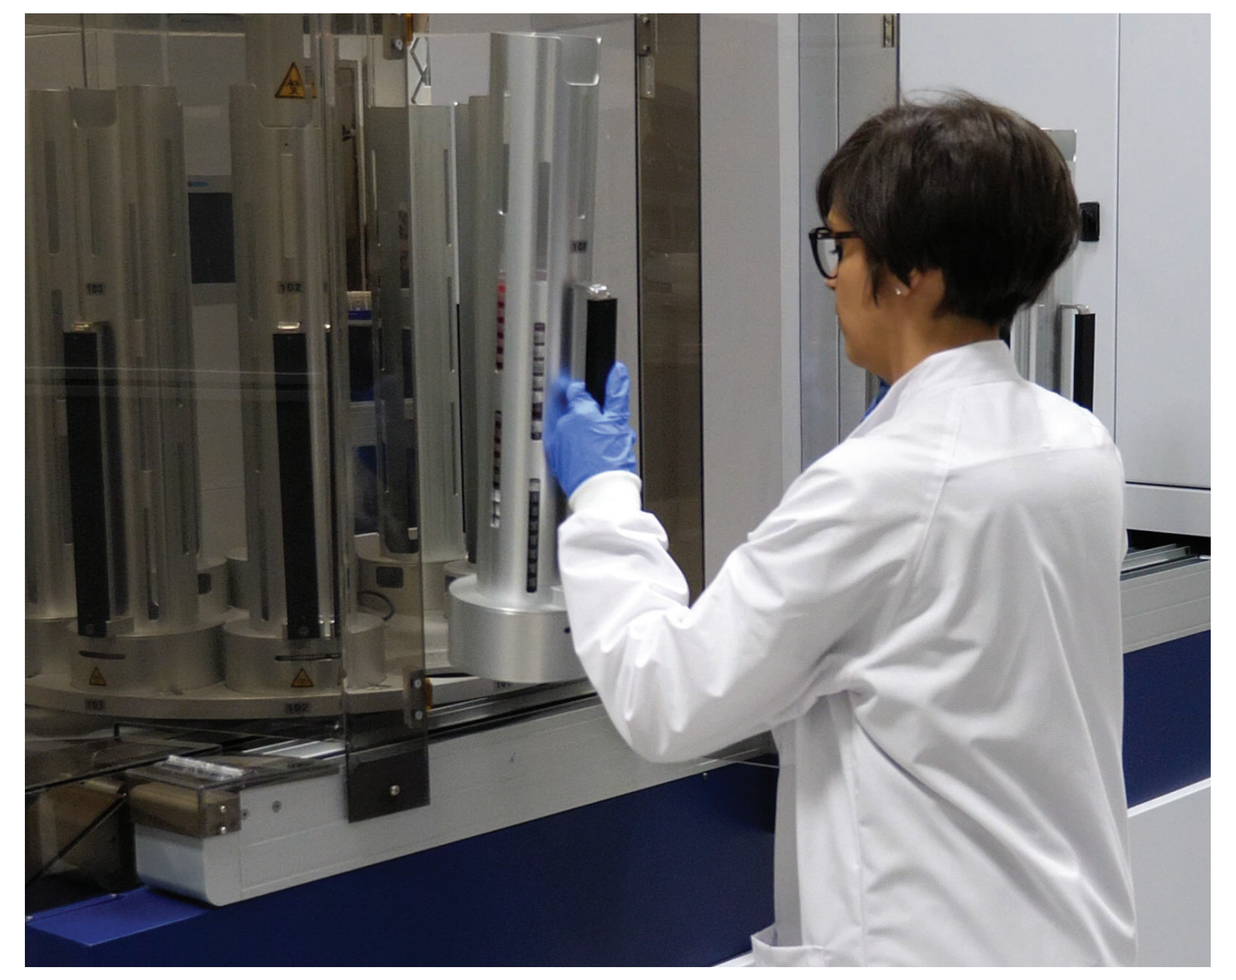

Figure 4: Colibrí samples the selected organisms from the culture plate.

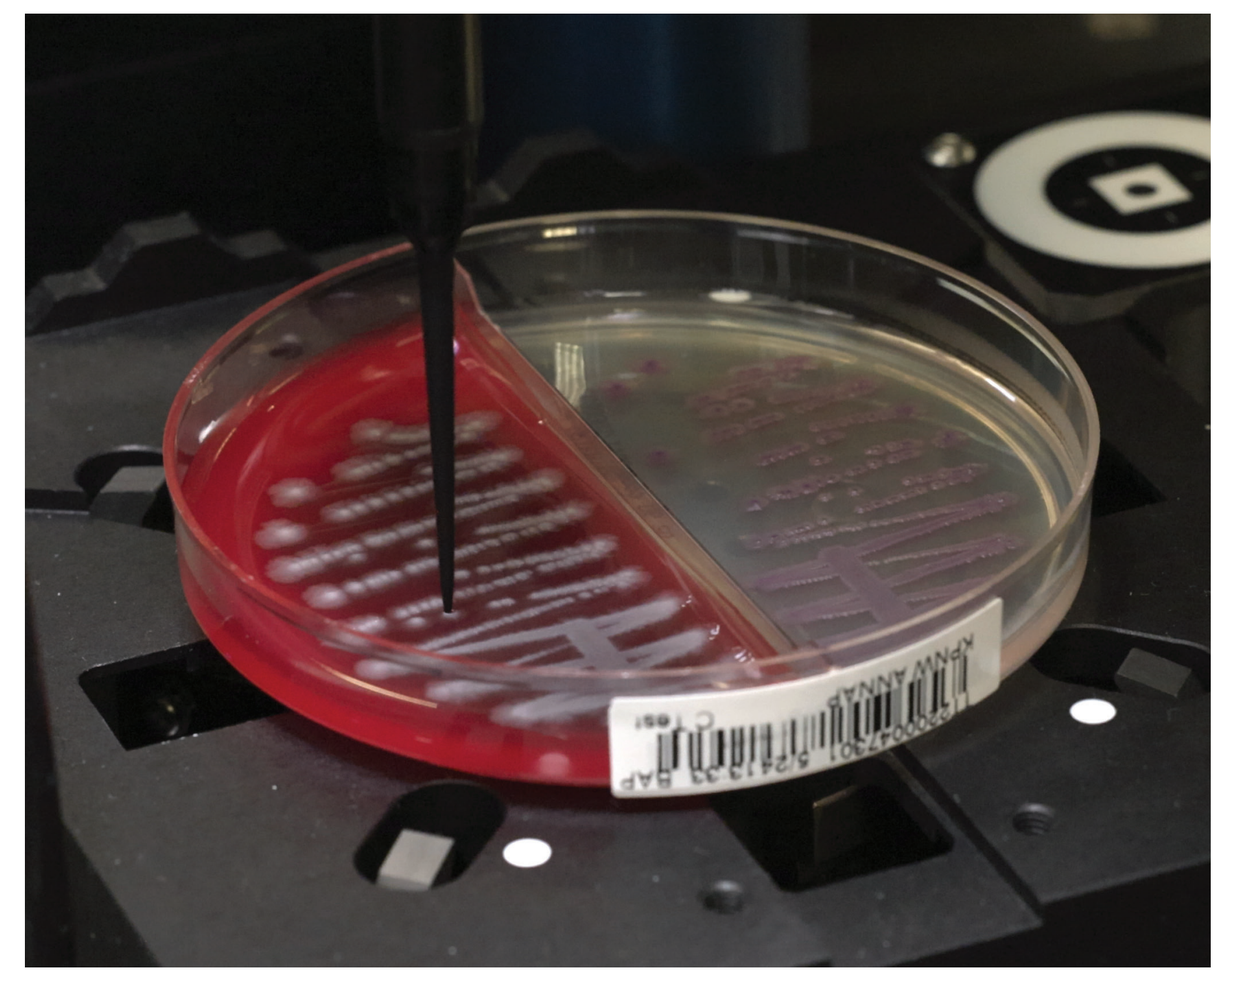

Figure 5: Colibrí places colonies in primary tube for the 0.5 McFarland turbidity and makes dilution tube for Vitek AST card.

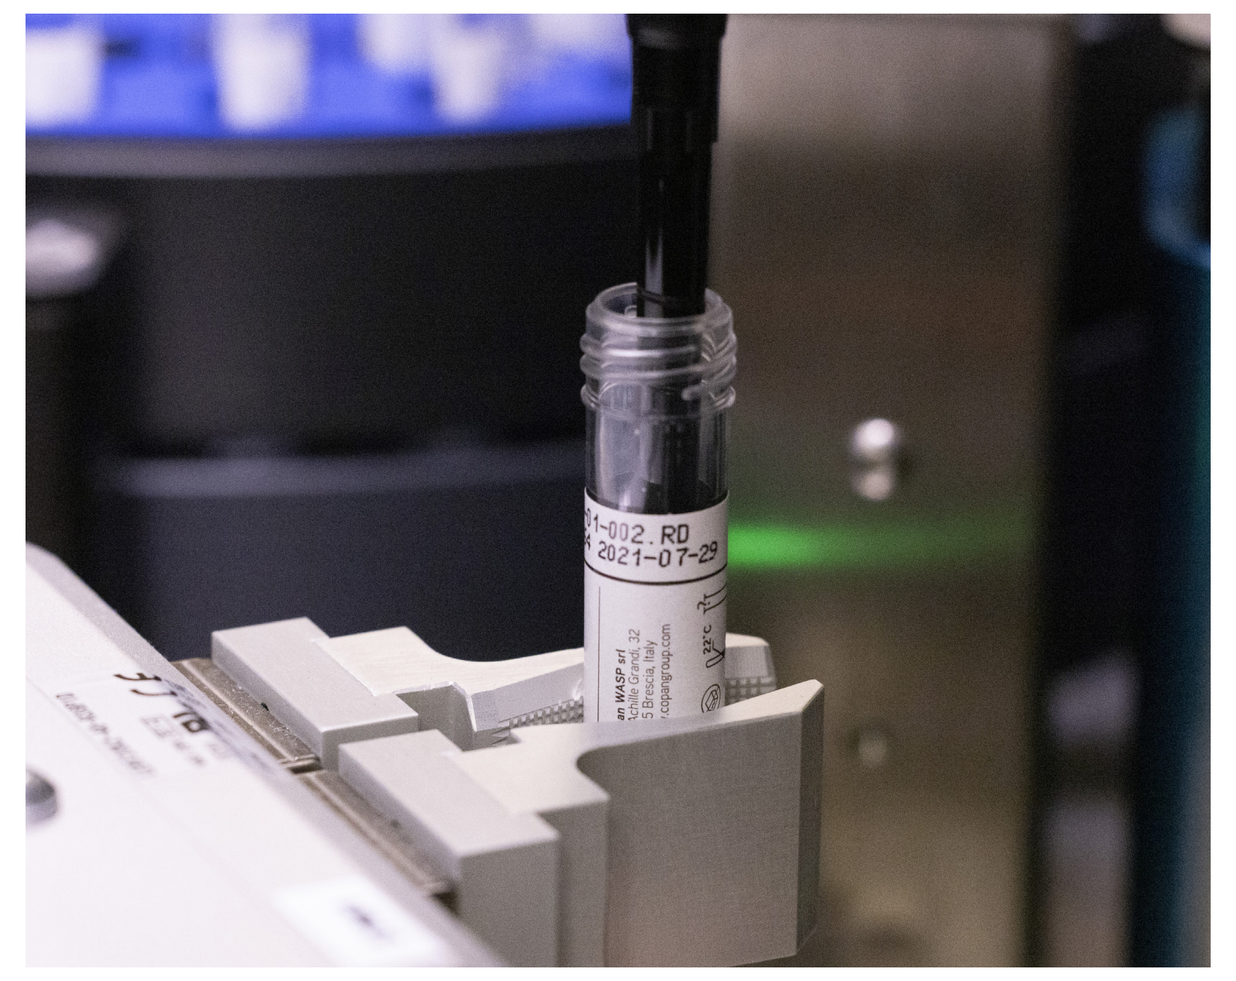

Figure 6: A purity plate was inoculated and streaked from each of the secondary tubes

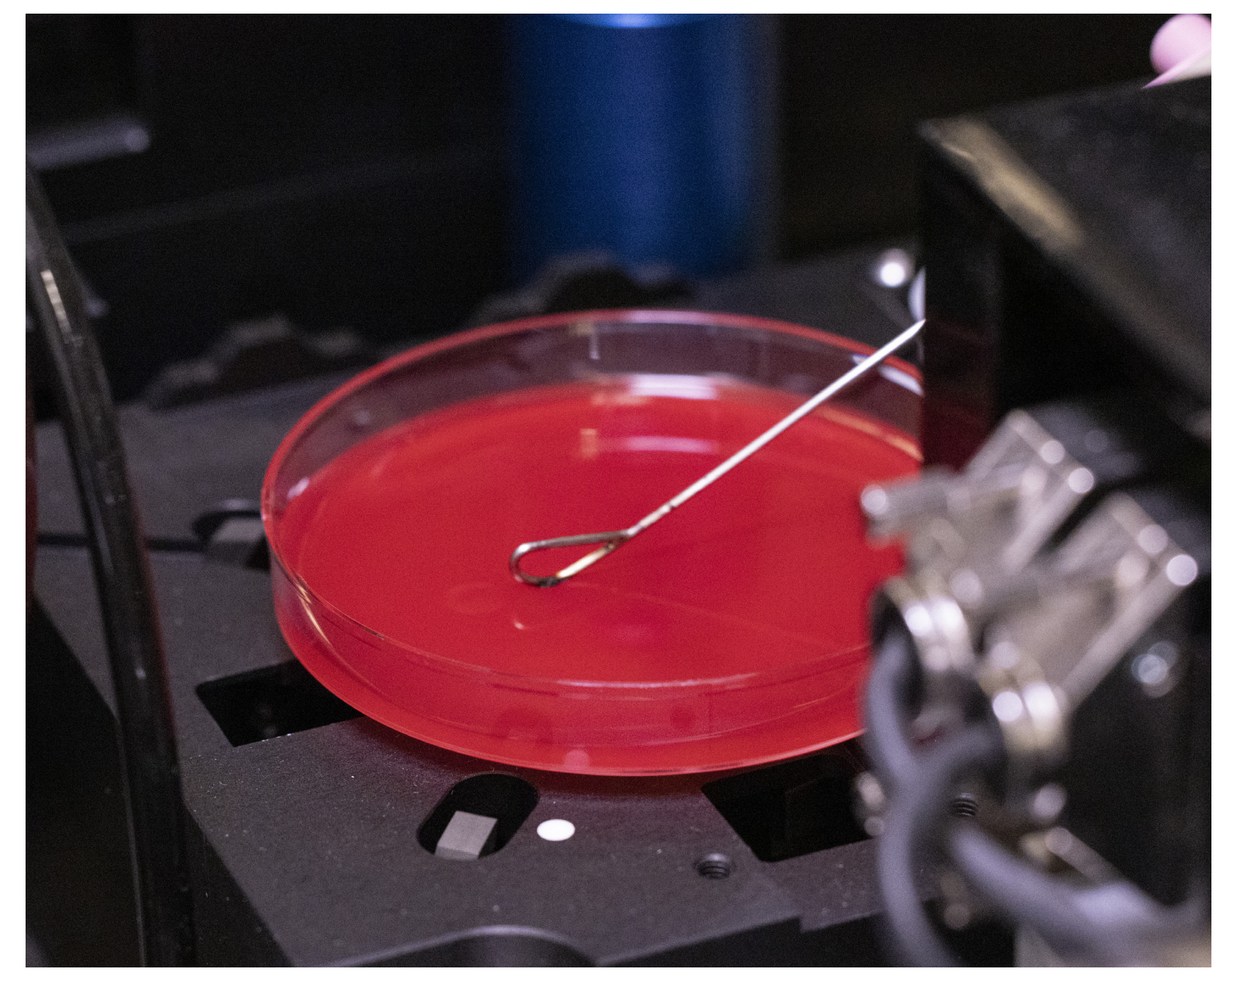

RESULTS:

A total of 1711 drug/organism were tested with 106 clinical isolates. The study compared 56 Gram negative organisms using the Vitek GN-81 card and 50 organisms were compared using the Vitek GP-75 card. The overall categorical agreement for the GN81 card was 99.5% all discordant results (N=3) were considered minor errors see **Table 1**. The overall agreement for the Vitek GP-75 card was 100% no categorical discordant results were found **Table 2**. There were 5 instances of > 1 dilution difference in MIC results between the methods: 1) cefazolin with a Klebsiella pneumoniae clinical isolate, 1) ampicillin and *Escherichia coli*, 1) erythromycin with a *Staphylococcus hominis* and 2) daptomycin with a *Staphylococcus aureus* clinical isolate for an overall +/- 1 dilution correlation of 99.99%.

Table 1: Comparison of methods for the Gram negative Vitek 2 GN-81

DRUG	COMPARISONS TESTED	NUMBER IN AGREEMENT	MINOR ERRORS	MAJOR ERROR	VERY MAJOR ERRORS	PERCENT AGREEMENT
Amikacin	56	56	0	0	0	100.0%
Amoxicillin/Clavulanate	48	47	1	0	0	98.0%
Ampicillin	29	29	0	0	0	100.0%
Cefazolin	55	55	0	0	0	100.0%
Cefepime	56	56	0	0	0	100.0%
Cefoxtin	49	49	0	0	0	100.0%
Ceftazidime	56	56	0	0	0	100.0%
Ceftriaxone	50	50	0	0	0	100.0%
Ciprofloxacin	56	56	0	0	0	100.0%
Ertapenem	42	42	0	0	0	100.0%
Gentamicin	56	55*	0	0	0	100.0%
Levofloxacin	56	56	0	0	0	100.0%
Meropenem	56	56	0	0	0	100.0%
Nitrofurantoin	49	47	2	0	0	96.0%
Piperacillin Tazobactam	54	54	0	0	0	100.0%
Tobramycin	56	56	0	0	0	100.0%
Tetracycline	49	49	0	0	0	100.0%
Sulfamethoxazole/ Trimethoprim	49	49	0	0	0	100.0%
TOTALS	**922**	**918**	**3**	**0**	**0**	**99.5%**

Table 2: Comparison of methods for the Gram positive Vitek 2 GP-75

DRUG	COMPARISONS TESTED	NUMBER IN AGREEMENT	MINOR ERRORS	MAJOR ERROR	VERY MAJOR ERRORS	PERCENT AGREEMENT
Cefoxtin Screen	24	24	0	0	0	100%
Ampicillin	11	11	0	0	0	100%
Ciprofloxacin	50	50	0	0	0	100%
Clindamycin	39	39	0	0	0	100%
Daptomycin	47	47	0	0	0	100%
Doxycycline	50	50	0	0	0	100%
Erythromycin	50	50	0	0	0	100%
Gentamicin	41	41	0	0	0	100%
Gentamicin Synergy	11	11	0	0	0	100%
Levofloxacin	50	50	0	0	0	100%
Linezolid	49	49	0	0	0	100%
Moxifloxacin	39	39	0	0	0	100%
Nitrofurantion	50	50	0	0	0	100%
Oxacillin	39	39	0	0	0	100%
Rifampicin	39	39	0	0	0	100%
Steptomycin Synergy	11	11	0	0	0	100%
Sulfamethoxazole/ Trimethoprim	39	39	0	0	0	100%
Tetracycline	50	50	0	0	0	100%
Tigecycline	50	50	0	0	0	100%
Vancomycin	50	50	0	0	0	100%
TOTALS	**789**	**789**	**0**	**0**	**0**	**100%**

CONCLUSIONS:

The Colibrí method for preparing suspensions for Vitek 2 antimicrobial susceptibility testing is equivalent to standard manual setup methods.

There was 99.75% categorical agreement and 99.99% agreement in MIC values (within +/-1 dilution) when comparing the Colibrí and the manual method for AST preparations.

The Colibrí provides an automated option for the highly repetitive task of Vitek AST setup freeing technologists time to perform other tasks in the laboratory.